Nintedanib plus docetaxel in lung adenocarcinoma patients following treatment with immune checkpoint inhibitors: preliminary efficacy and safety results of the non-interventional study VARGADO

Poster #397
Abstract 9074

Christian Grohé,[1] Wolfgang Blau,[2] Wolfgang Gleiber,[3] Siegfried Haas,[4] Harald Müller-Huesmann,[5] Mathias Schulze,[6] Judith Atz,[7] Rolf Kaiser[8]

[1]Department of Pneumology, ELK, Berlin, Germany; [2]Department of Internal Medicine V, University Hospital Giessen, Giessen, Germany; [3]University Hospital Frankfurt, Pneumology/Allergology, Frankfurt, Germany; [4]Friedrich-Ebert Hospital Neumuenster, Clinics for Haematology, Oncology and Nephrology, Neumuenster, Germany; [5]Klinik für Hämatologie und Onkologie, Bruederkrankenhaus St. Josef, Paderborn, Germany; [6]Praxis Dr. Schulze, Zittau, Germany; [7]Medical Affairs Oncology, Boehringer Ingelheim Pharma GmbH & Co KG, Ingelheim Am Rhein, Germany; [8]Boehringer Ingelheim Pharma GmbH & Co, KG, Germany and Institute of Pharmacology, Johannes Gutenberg-University Mainz, Germany

INTRODUCTION

- Nintedanib is an oral, triple angiokinase inhibitor targeting vascular endothelial growth factor (VEGF) receptors 1–3, platelet-derived growth factor receptors (PDGF) α/β, and fibroblast growth factor (FGF) receptors 1–3,[1] as well as RET[2]
- Nintedanib is approved in the EU and other countries in combination with docetaxel for the treatment of patients with locally advanced, metastatic or locally recurrent non-small cell lung cancer (NSCLC) of adenocarcinoma histology after first-line chemotherapy[3]
- This approval was supported by the Phase III LUME-Lung 1 trial, in which nintedanib plus docetaxel significantly prolonged progression-free survival (PFS) versus placebo plus docetaxel in patients with NSCLC who had progressed on first-line chemotherapy; overall survival (OS) was also significantly longer in the nintedanib plus docetaxel arm in patients with adenocarcinoma histology[4]
- Immune checkpoint inhibitors (ICIs) are a new standard of care in advanced non-targetable NSCLC,[5] but the optimal treatment sequence after progression on ICI ± chemotherapy remains to be established
- Understanding the underlying tumor biology may help to guide the selection of the optimal treatment sequence. As well as promoting angiogenesis, excessive VEGF can also create an immunosuppressive tumor microenvironment (TME) by modulating the function of immune cells and restricting their migration into the tumor.[6,7] These mechanisms are likely to contribute to ICI resistance, and, conversely, would prime the tumor for anti-angiogenic therapy
- We hypothesize that an anti-angiogenic treatment strategy involving inhibition of VEGF, as well as PDGF and FGF, could support vessel normalization and improve access of immune cells to the tumor, tipping the balance towards an immunosupportive TME in an 'angio-immunogenic switch' (**Figure 1**)

Figure 1. A hypothesized 'angio-immunogenic switch'

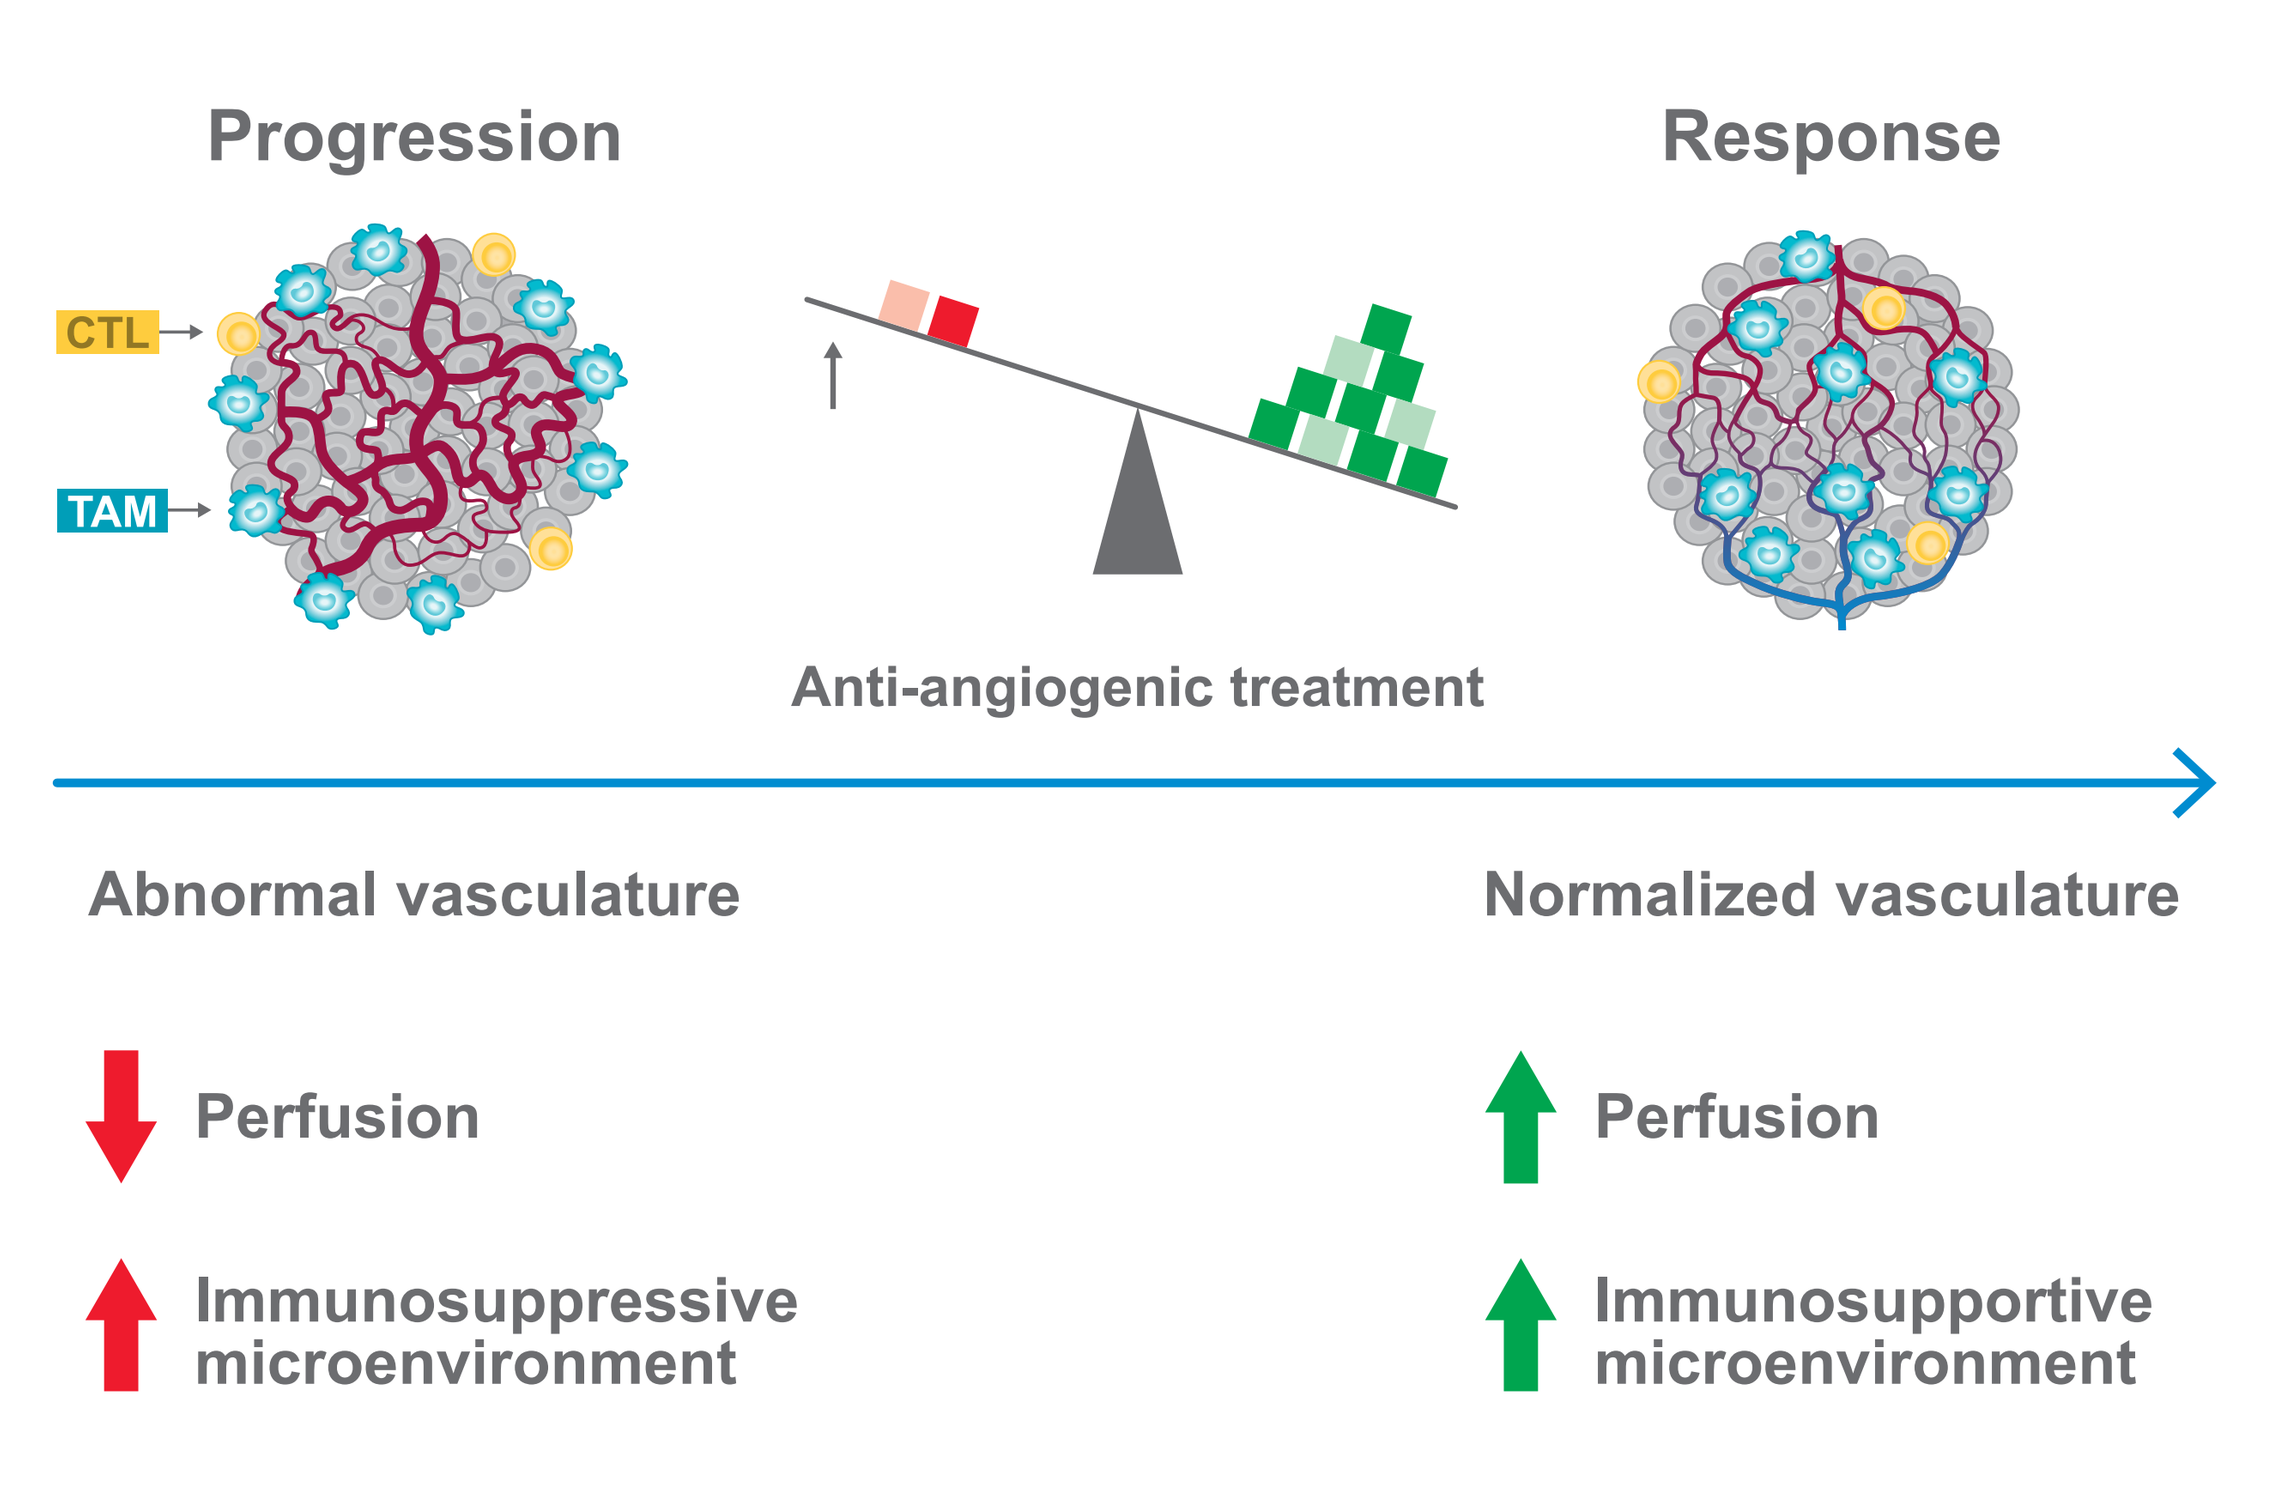
Progression
Response
CTL
TAM
Anti-angiogenic treatment
Abnormal vasculature
Normalized vasculature
Perfusion
Perfusion
Immunosuppressive microenvironment
Immunosupportive microenvironment

Modified from Fukumura D, et al. Nat Rev Clin Oncol 2018;15:325–40.
CTL, cytotoxic T lymphocyte; TAM, tumor-associated macrophage.

- Only a few clinical datasets have been published to guide treatment decisions after progression on ICIs[8,9]
- Here, we present updated data from a cohort of patients who received nintedanib plus docetaxel after chemotherapy and ICI therapy, as part of the non-interventional, prospective VARGADO[10] study

STUDY DESIGN AND PATIENT POPULATION

- VARGADO is a prospective, non-interventional study of nintedanib plus docetaxel after first-line chemotherapy in the routine clinical treatment of patients with locally advanced, metastatic or locally recurrent adenocarcinoma NSCLC[10]
- Three patient cohorts in VARGADO are being evaluated (**Figure 2**)
- Between 15 March 2015 and 30 April 2019, 345 patients have been enrolled in centers across Germany
- We present an updated interim analysis of Cohort B (N=25)

Figure 2. Patient cohorts in VARGADO

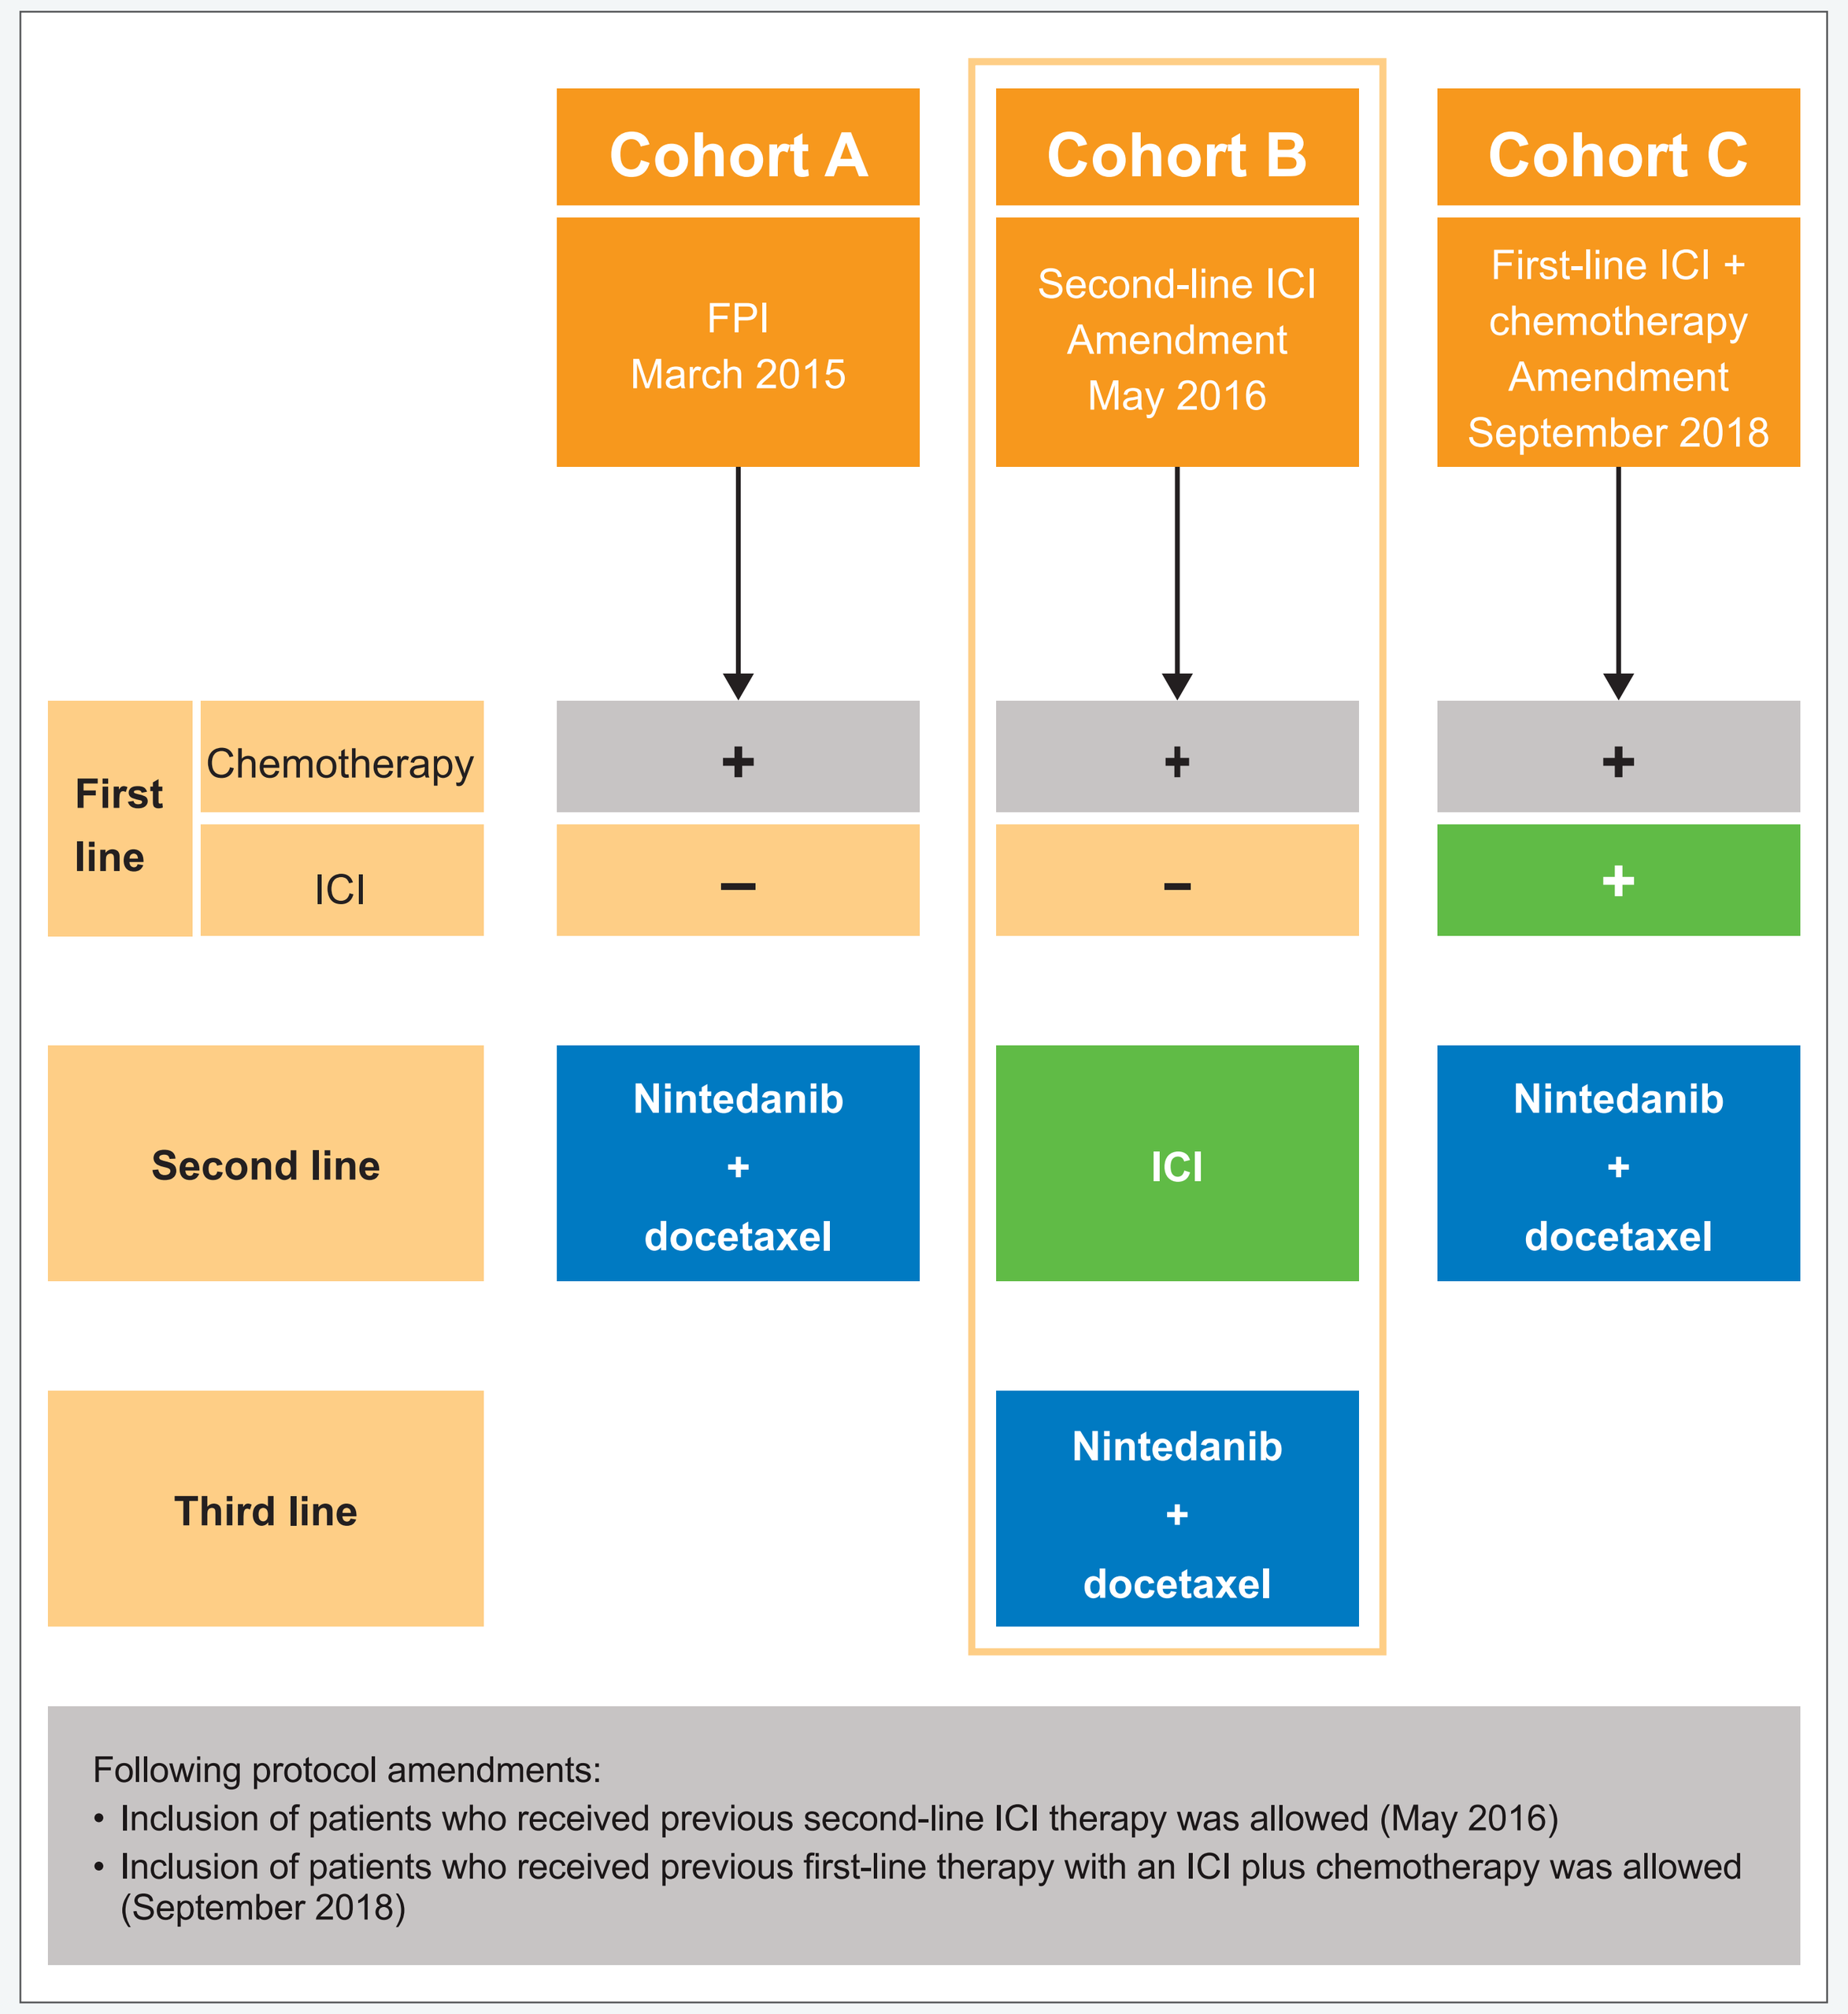
Cohort A
FPI
March 2015
Cohort B
Second-line ICI
Amendment
May 2016
Cohort C
First-line ICI +
chemotherapy
Amendment
September 2018
First line
Chemotherapy
ICI
+
–
+
–
+
+
Second line
Nintedanib + docetaxel
ICI
Nintedanib + docetaxel
Third line
Nintedanib + docetaxel
Following protocol amendments:
• Inclusion of patients who received previous second-line ICI therapy was allowed (May 2016)
• Inclusion of patients who received previous first-line therapy with an ICI plus chemotherapy was allowed (September 2018)

FPI, first patient in; ICI, immune checkpoint inhibitor.

- Nintedanib and docetaxel were administered according to the approved label
- Patients were followed up for safety and efficacy for up to 24 months after the start of treatment. Patient data were collected during routine clinic visits
- All patients provided signed informed consent
- The primary endpoint is OS rate 1 year after the start of treatment with nintedanib plus docetaxel. Secondary endpoints include PFS, OS, objective response rate and disease control rate
- Incidence and severity of adverse events (AEs) were reported according to National Cancer Institute Common Terminology Criteria for Adverse Events, version 4.03

RESULTS

Patient characteristics

- Baseline characteristics for the 25 patients included in this analysis are shown in **Table 1**

Table 1. Baseline characteristics (N=25)

Median age, years (range)		59 (45–76)
Sex, n (%)	Male	17 (68)
ECOG PS, n (%)	0	4 (16)
	1	13 (52)
	2	2 (8)
	3	1 (4)
	Missing	5 (20)
Tumor stage at baseline, n (%)	IV	13 (52)
	<IV	6 (24)
	Unknown	4 (16)
	Missing	2 (8)
Current or former smokers, n (%)		24 (96)
Brain metastases, n (%)	Yes	6 (24)
Previous first-line therapy, n (%)	Pemetrexed	18 (72)
	Cisplatin	15 (60)
	Carboplatin	12 (48)
	Bevacizumab	8 (32)
	Vinorelbine	4 (16)
	Paclitaxel	2 (8)
	Docetaxel	1 (4)
Previous second-line therapy, n (%)	Nivolumab	18 (72)
	Pembrolizumab	6 (24)
	ICI not yet documented	1 (4)
Best response to second-line ICI therapy, n (%)	Complete response	1/14 (7)
	Partial response	2/14 (14)
	Stable disease	2/14 (14)
	Progressive disease	9/14 (64)
	Not yet documented	11

ECOG PS, Eastern Cooperative Oncology Group performance status; ICI, immune checkpoint inhibitor.

Efficacy

- At the time of this interim analysis (data cut-off: 4 December 2018), median duration of follow-up was 7.9 months for patients treated with nintedanib plus docetaxel (95% confidence interval [CI]: 3.5–12.2)
- Sixteen PFS events had occurred (12 patients had disease progression and four patients died). Eight patients were censored and data were not yet available for one patient
- Median PFS was 5.5 months (95% CI: 2.5–8.2; **Figure 3**) in the third-line setting when patients were treated with nintedanib plus docetaxel after failure of second-line ICI therapy

Figure 3. PFS by investigator assessment (n=24)

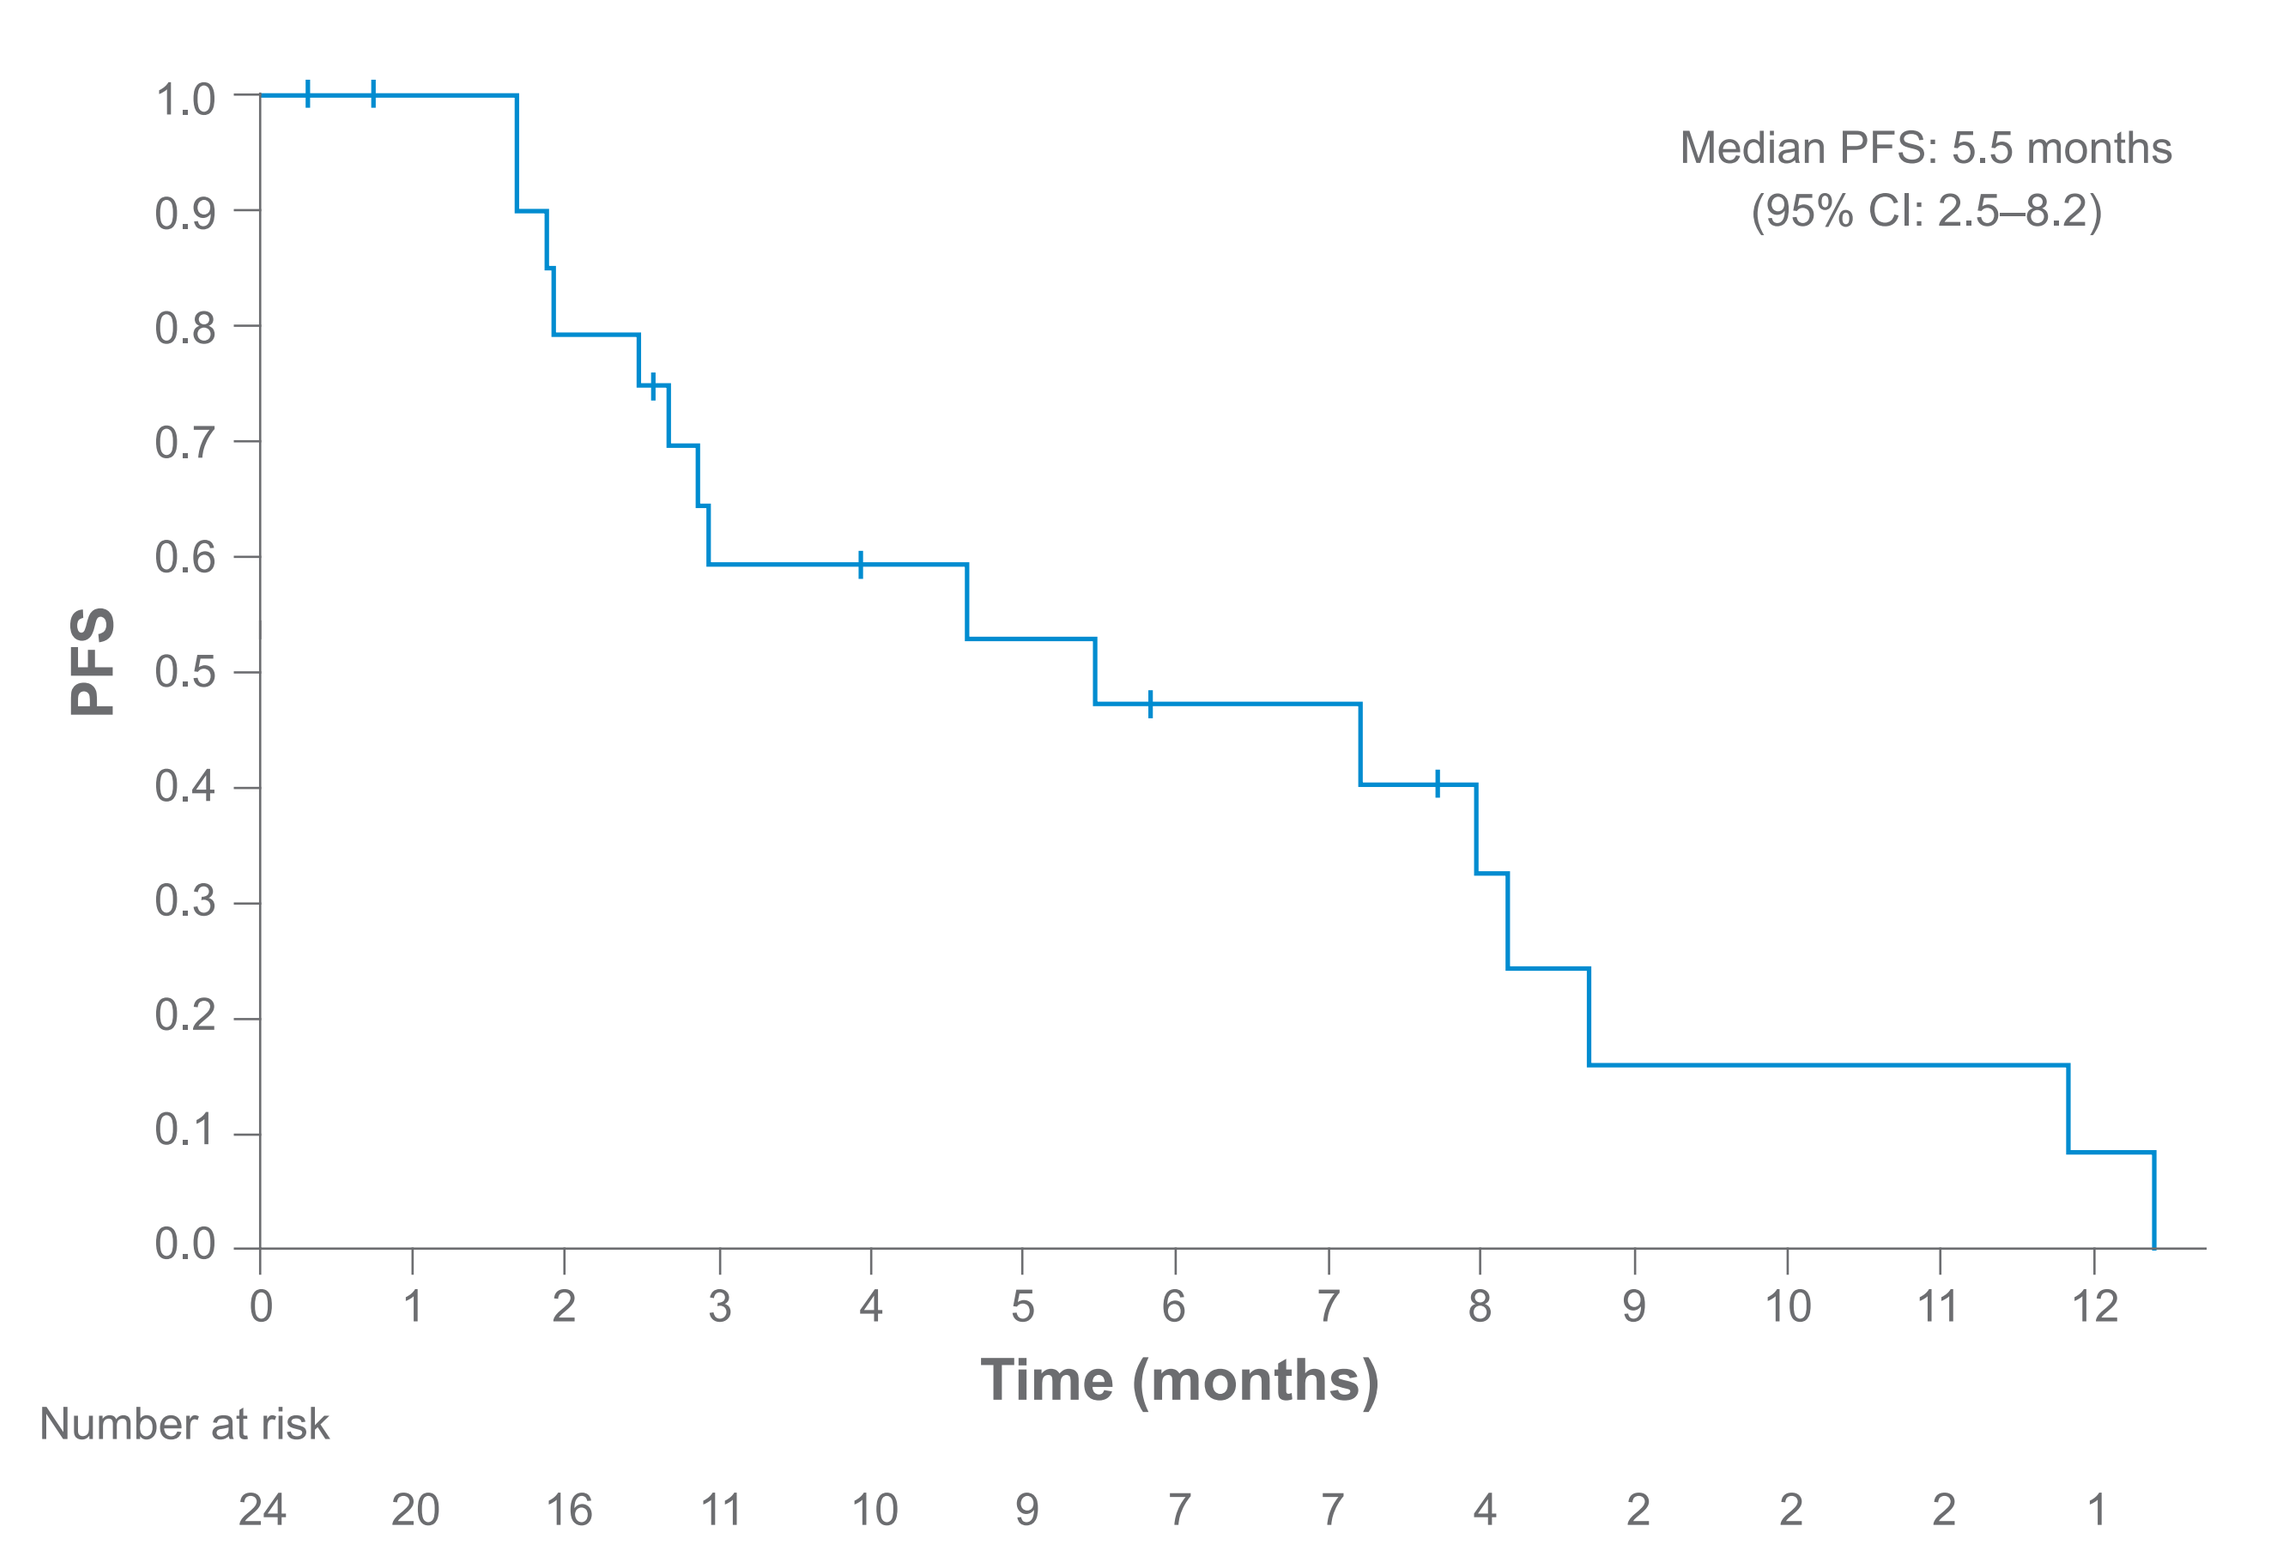
Median PFS: 5.5 months
(95% CI: 2.5–8.2)
PFS
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
0 1 2 3 4 5 6 7 8 9 10 11 12
Time (months)
Number at risk
24 20 16 11 10 9 7 7 4 2 2 2 1

Data cut-off: 4 December 2018.
CI, confidence interval; PFS, progression-free survival.

Figure 4. Swimmer plot showing treatment duration by treatment for each patient with a documented response (n=20)

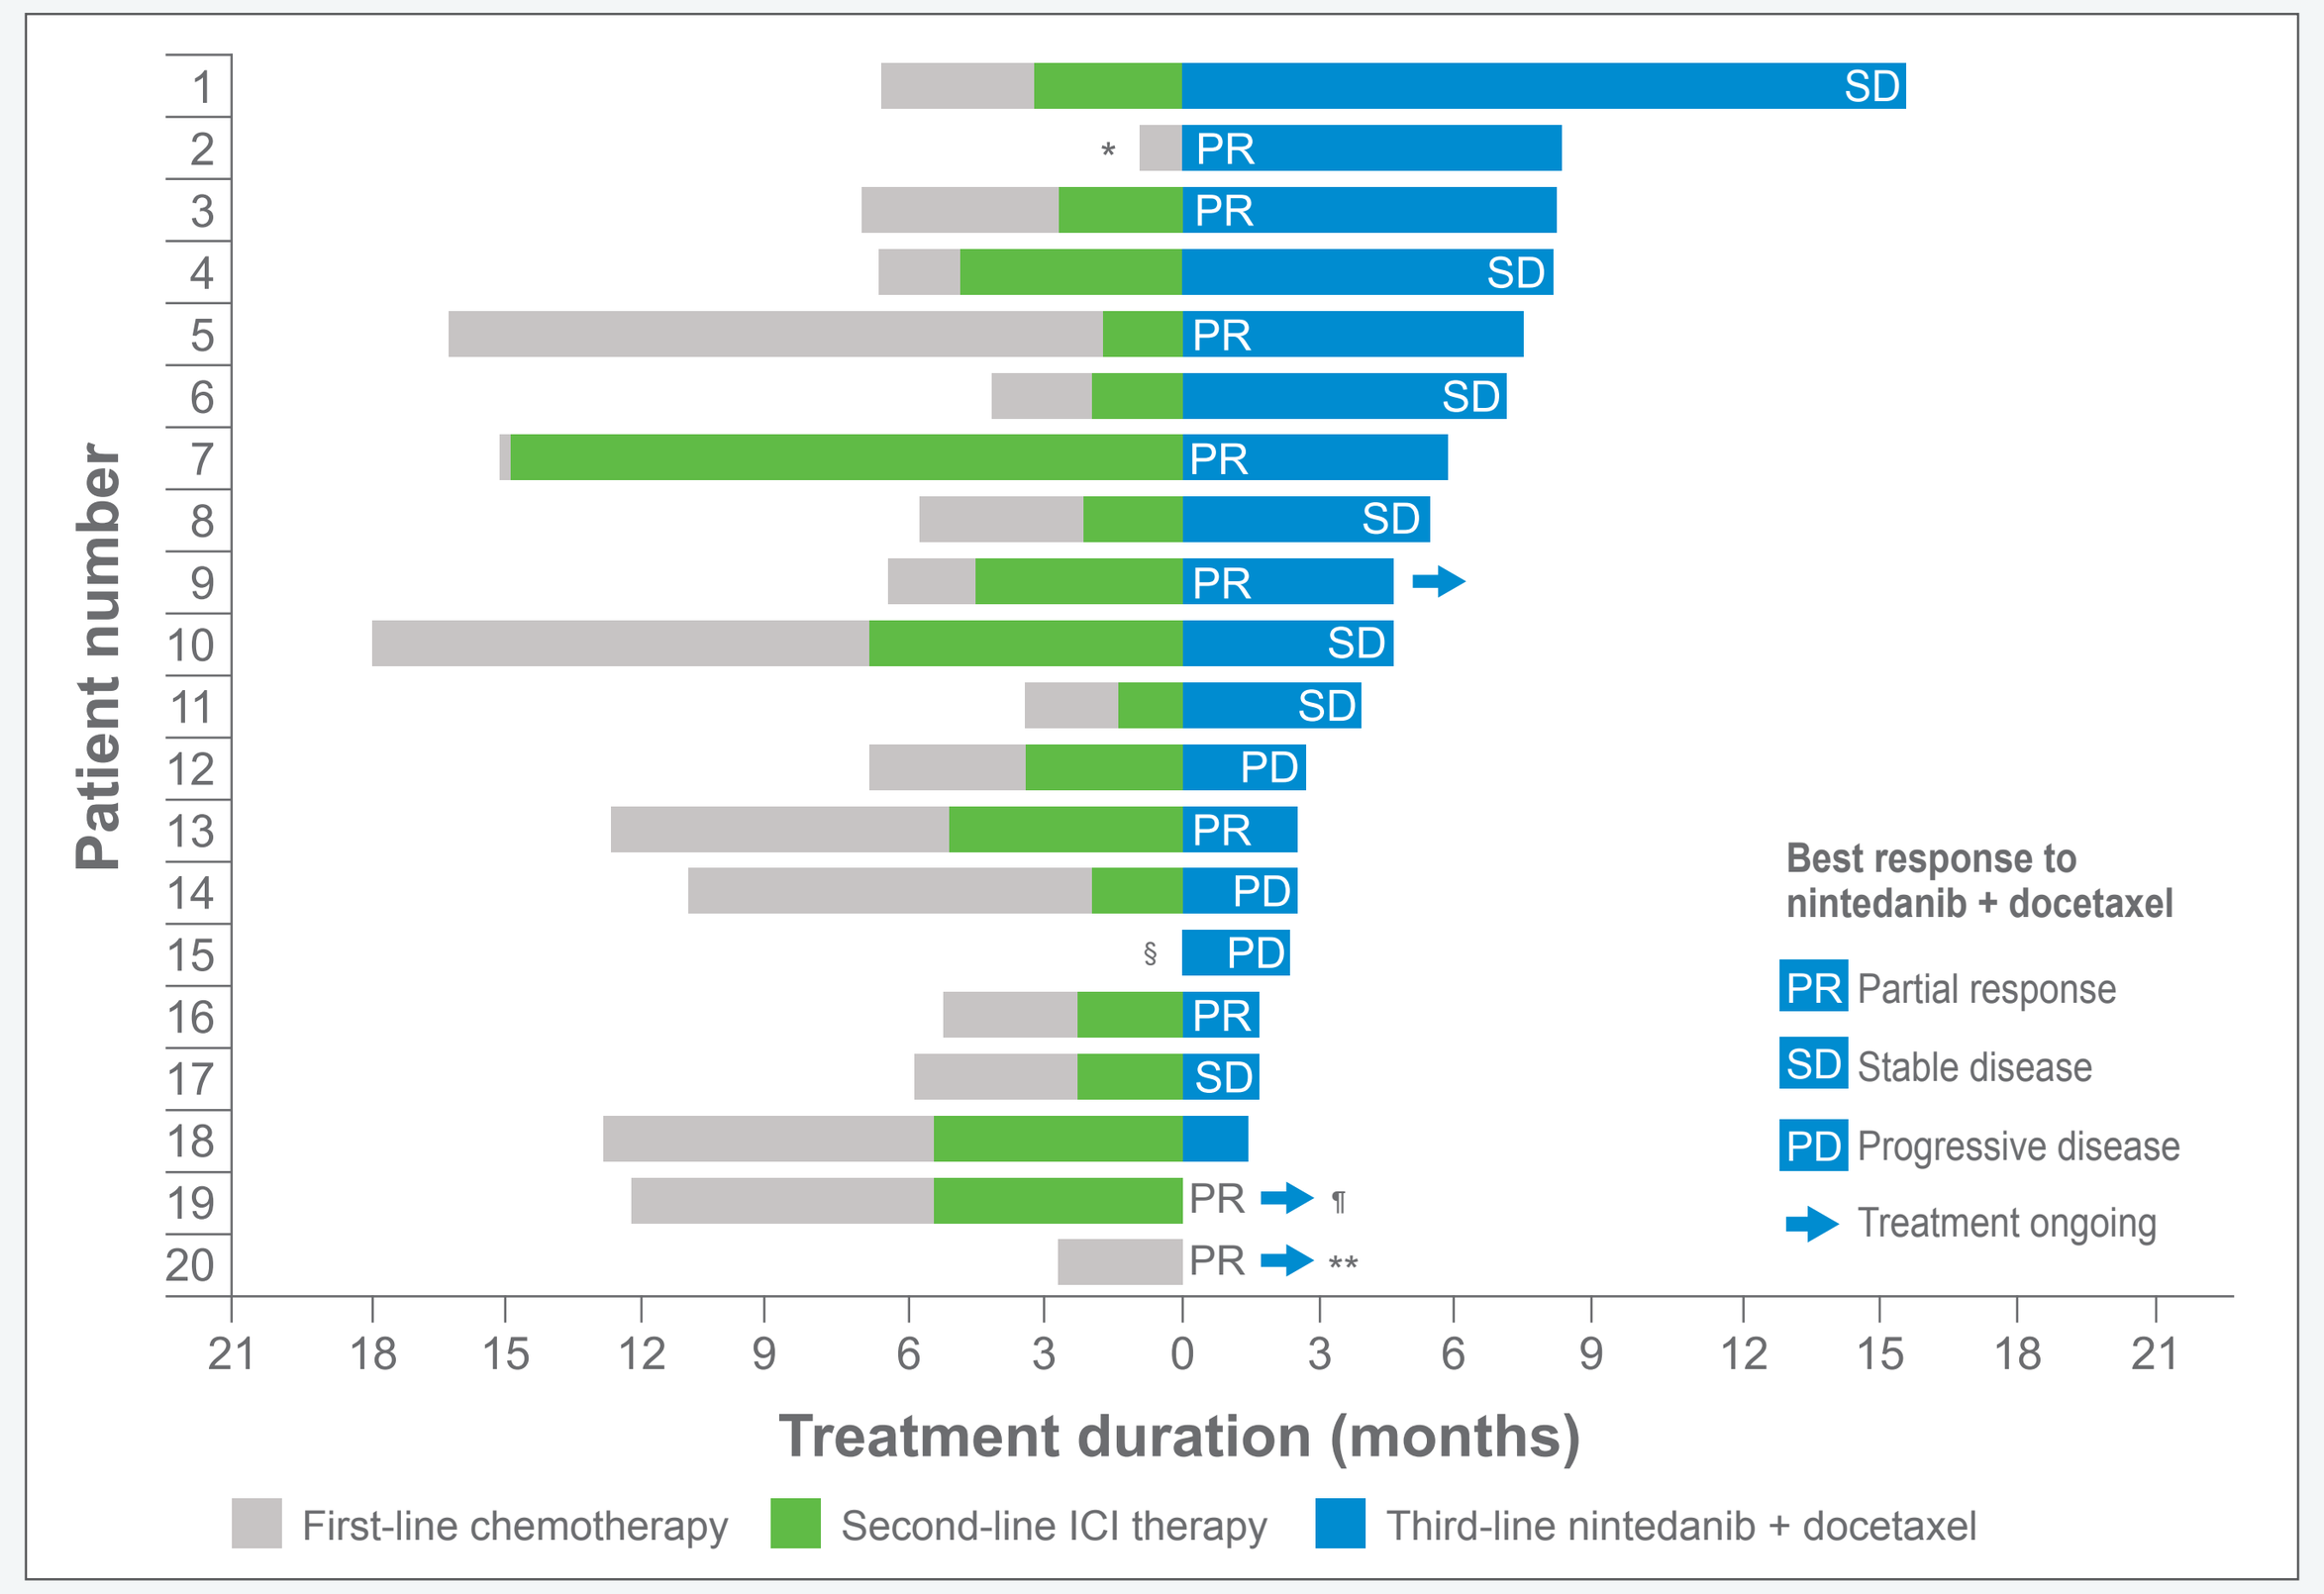
Patient number
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20
SD
PR
PR
SD
PR
SD
PR
SD
PR
SD
SD
PD
PR
PD
PD
PR
SD
PR
PR
*
§
¶
**
Best response to nintedanib + docetaxel
PR Partial response
SD Stable disease
PD Progressive disease
Treatment ongoing
21 18 15 12 9 6 3 0 3 6 9 12 15 18 21
Treatment duration (months)
First-line chemotherapy
Second-line ICI therapy
Third-line nintedanib + docetaxel

*Time on chemotherapy not yet documented; five chemotherapy cycles documented; §Time on chemotherapy and ICI therapy not yet documented; four chemotherapy cycles documented; ¶Clinically documented response; time on nintedanib plus docetaxel not yet documented; **Clinically documented response; time on ICI therapy and nintedanib plus docetaxel not yet documented.
Data cut-off: 4 December 2018.
ICI, immune checkpoint inhibitor.

- OS data are not yet mature and are not reported here
- At the time of analysis, objective response rate and disease control rate data were available for 20 patients who received third-line nintedanib plus docetaxel after failure of ICI therapy (**Table 2**)
 - The partial response rate was 9/20 (45%)
 - The disease control rate was 16/20 (80%)

Table 2. Best response to third-line nintedanib plus docetaxel after failure of ICI therapy (n=20)

Objective response rate, n (%)	9 (45)
Complete response, n (%)	0
Partial response, n (%)	9 (45)
Stable disease, n (%)	7 (35)
Disease control rate, n (%)	16 (80)
Progressive disease, n (%)	4 (20)

Data cut-off: 4 December 2018.

Safety

- Safety was evaluated in all 25 patients
- 92% of patients had AEs, with 72% experiencing drug-related AEs
- Nine patients (36%) had at least one nintedanib dose reduction and five patients (20%) had at least one docetaxel dose reduction
- Grade ≥3 treatment-emergent AEs occurred in 60% of patients; serious treatment-emergent AEs occurred in 52% of patients
- Treatment-emergent AEs led to discontinuation of study treatment in 36% of patients
- The most common treatment-related AEs are shown in **Table 3**

Table 3. Drug-related AEs reported in >10% of patients (N=25)

	Nintedanib related		Docetaxel related	
	All grades n (%)	Grade ≥3 n (%)	All grades n (%)	Grade ≥3 n (%)
Diarrhea	9 (36)	0	8 (32)	0
Stomatitis	6 (24)	3 (12)	6 (24)	3 (12)
Decreased white blood cell count	3 (12)	3 (12)	6 (24)	5 (20)
Nausea	3 (12)	1 (4)	4 (16)	2 (8)
Fatigue	1 (4)	0	4 (16)	1 (4)
Deterioration in general physical health	2 (8)	0	3 (12)	1 (4)
Decreased appetite	1 (4)	1 (4)	3 (12)	1 (4)

Data cut-off: 4 December 2018.
AE, adverse event.

CONCLUSIONS

- These data provide initial clinical evidence that patients who progressed on previous ICI therapy may benefit from treatment with nintedanib plus docetaxel
- Results show consistent evidence of the clinical benefit of third-line nintedanib plus docetaxel in this setting in terms of PFS, response rate and disease control rate
- We propose that the rational sequencing of anti-angiogenic therapy after ICIs may be a promising approach to treatment in this patient population that warrants further investigation

REFERENCES

1. Hilberg F, et al. Cancer Res 2008;68:4774–82.
2. Boehringer Ingelheim. Data on file.
3. Vargatef® Summary of Product Characteristics. http://www.ema.europa.eu/docs/en_GB/document_library/EPAR_-_Product_Information/human/002569/WC500179970.pdf (Accessed: 7 May 2019).
4. Reck M, et al. Lancet Oncol 2014;15:143–55.
5. Planchard D, et al. Ann Oncol 2018;29(Suppl. 4):iv192–237.
6. Fukumura D, et al. Nat Rev Clin Oncol 2018;15:325–40.
7. van der Woude LL, et al. Trends Cancer 2017;3:797–808.
8. Molife C, et al. Future Oncol 2019. doi: 10.2217/fon-2018-0876. [Epub ahead of print].
9. Corral J, et al. Clin Transl Oncol 2019. doi: 10.1007/s12094-019-02053-7. [Epub ahead of print].
10. Grohé C, et al. Ann Oncol 2018;29(Suppl. 10): Abstract 55P.

ACKNOWLEDGEMENTS

Disclosures: The authors were fully responsible for all content and editorial decisions, were involved in all stages of poster development and have approved the final version.
During the preparation of this poster, medical writing assistance, supported financially by Boehringer Ingelheim, was provided by Syneos Health, UK.
Disclaimer: Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without written permission from ASCO® and the author of this poster.
To download a copy of the poster, scan the QR code or type the following URL into your browser: https://bit.ly/2YGoINZ
Corresponding author: Christian Grohé (Christian.Grohe@pgdiakonie.de).

Presented at the American Society of Clinical Oncology (ASCO) Annual Meeting, Chicago, USA, 31 May–4 June 2019

VGTO232-06-2019